Complete and specific regression of disseminated tumors in a novel rat mesenchymal three-tumor model after intralesional treatment with the nonapeptide LTX-315 (Oncopore™)

Ø. REKDAL[1], J. NESTVOLD[3], M. WANG[2], K. CAMILIO[4], B. SVEINBJØRNSSON[4], G. KVALHEIM[2].
[2]The Norwegian Radium Hospital, [3]University of Oslo, [4]University of Tromsø, [1]Lytix Biopharma AS, NO-9294 Tromsø, Norway.

Lytix Biopharma

Background

Intralesional administration of anticancer drugs derived from host defense peptides represents a novel innovative immunotherapeutic strategy.

Our structure-activity relationship studies on host defense peptides have culminated in the engineering of small peptides with enhanced anticancer activity (1). Ultimately, these efforts have led to the development of a chemically modified 9-mer peptide, LTX-315.

LTX-315 has shown to induce complete tumor regression and prevent relapse and metastasis in several animal models (2,3).
A phase 1 study has been completed with LTX-315 and a new clinical study is ongoing.

Aim

To investigate whether LTX-315 induces abscopal effects in a novel rat mesenchymal three-tumor model.

Results

Fig. 1 LTX-315 induces the release of DAMPs in vitro

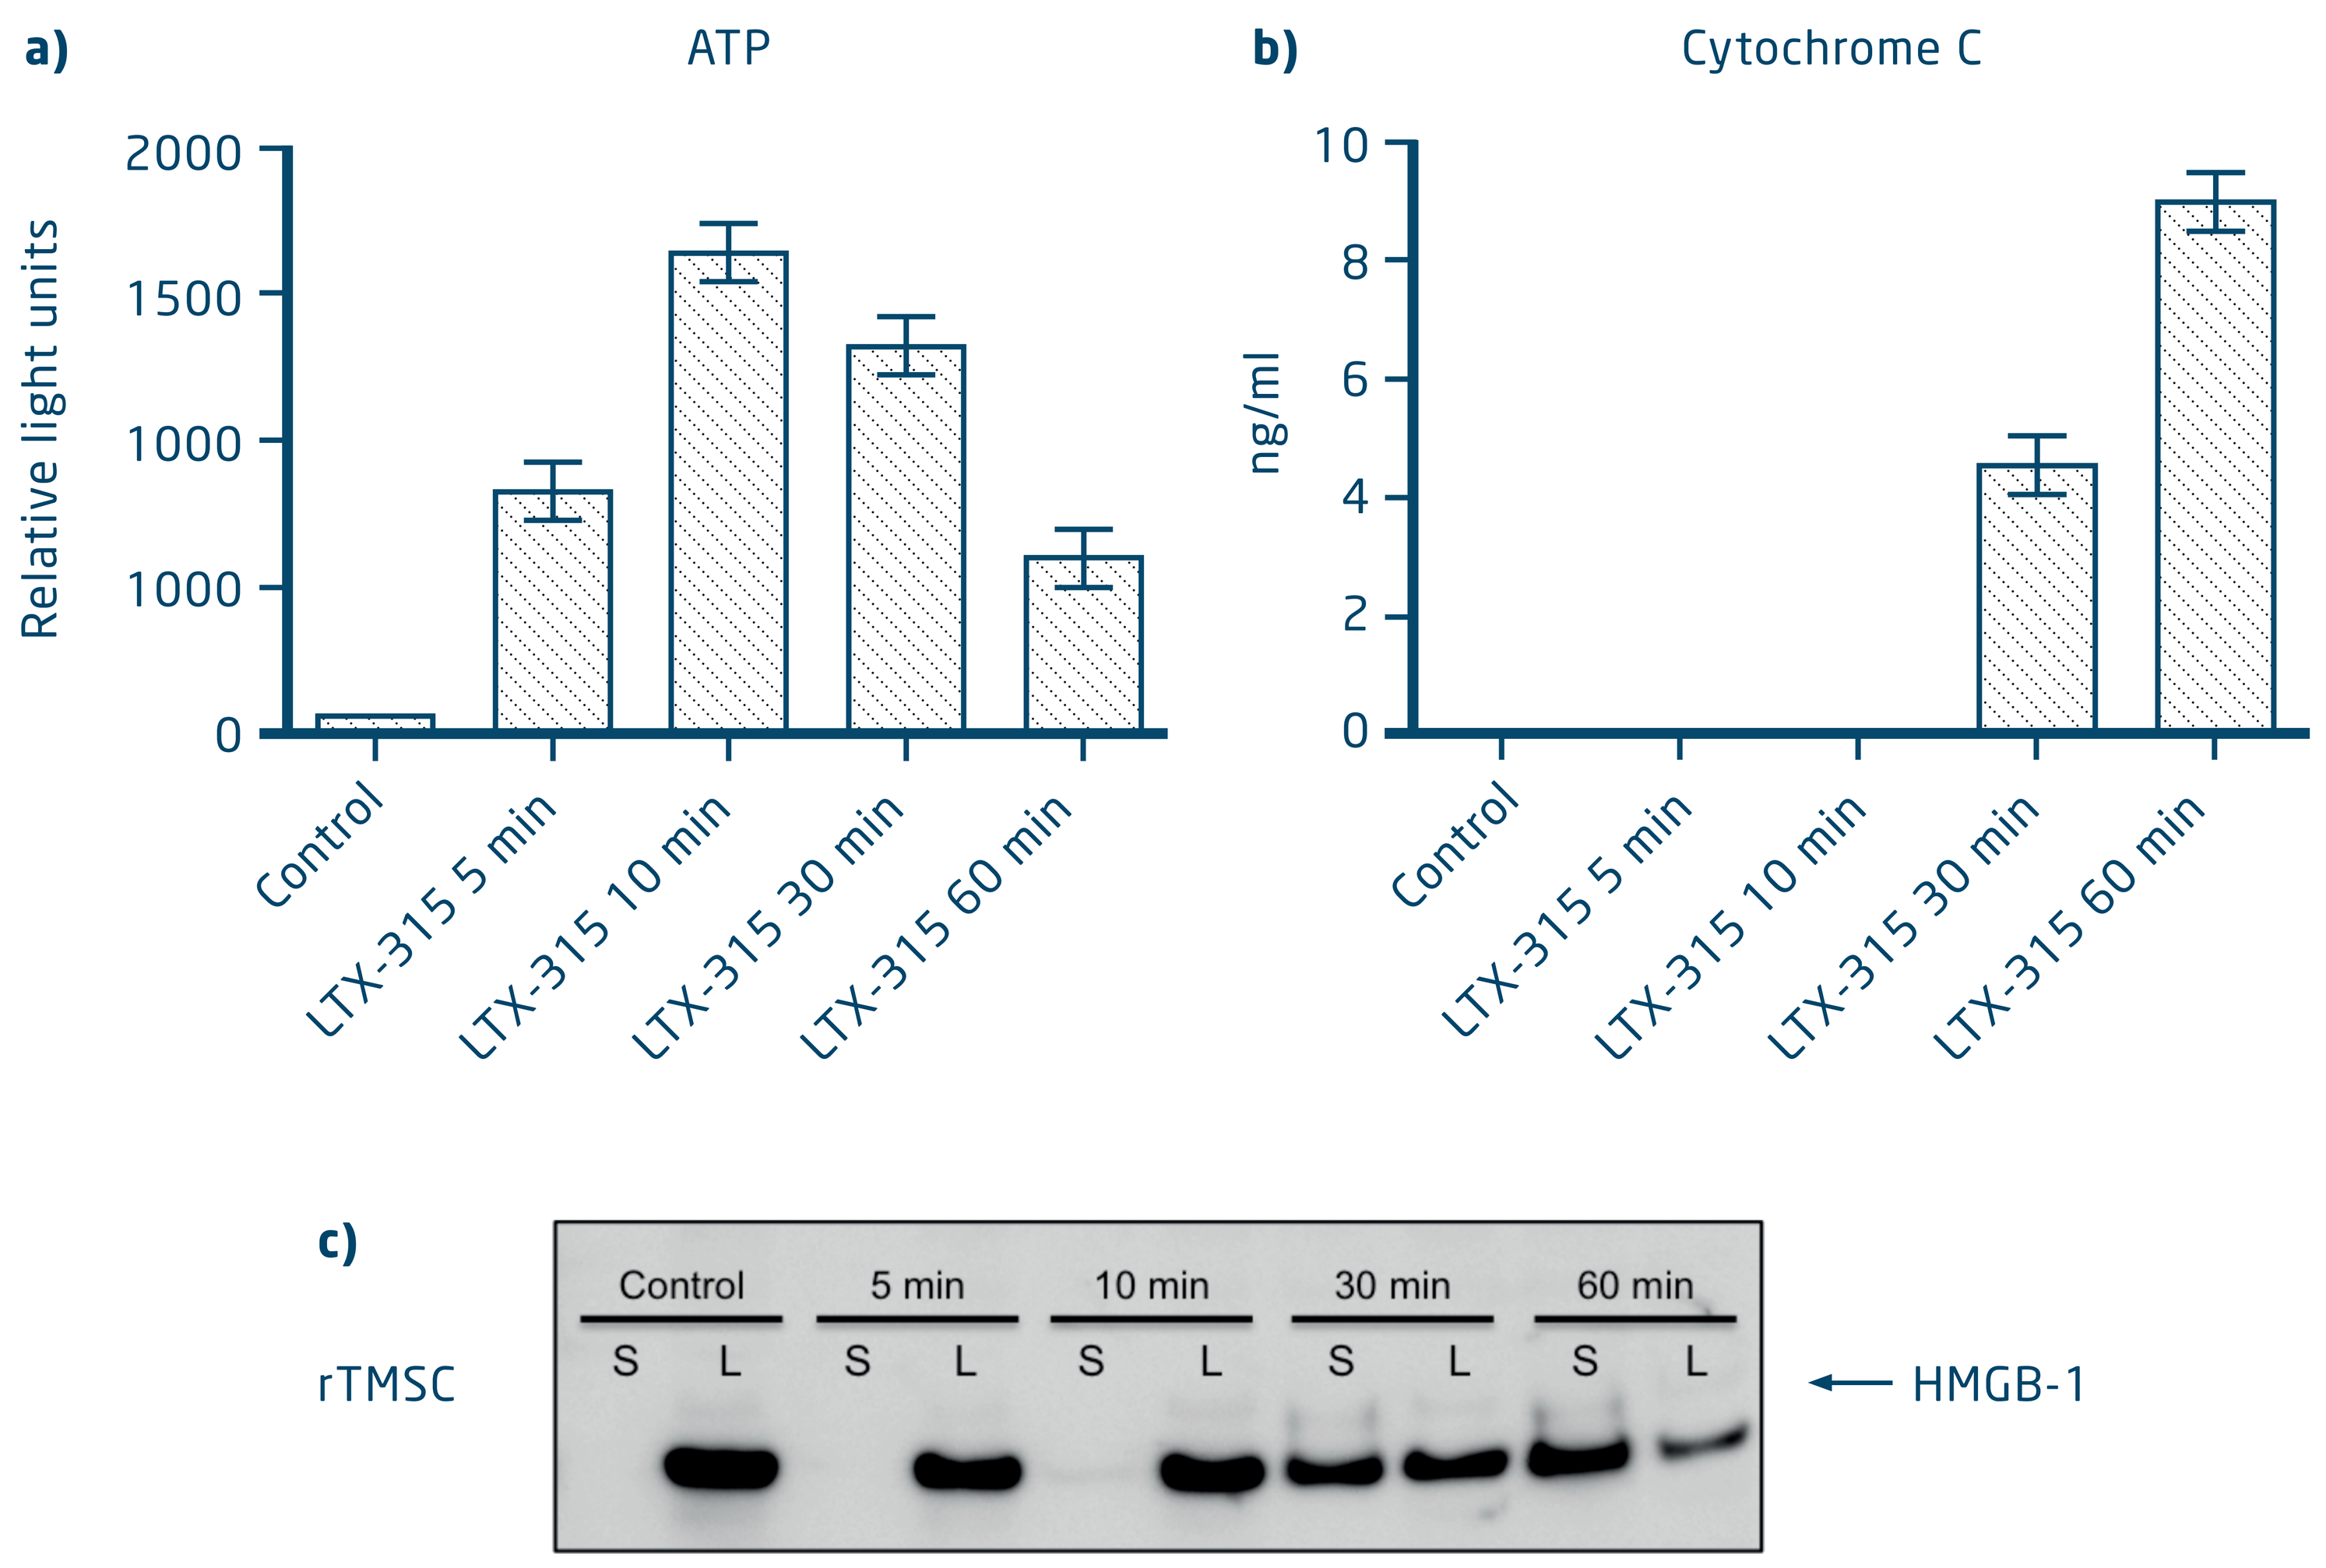
a)
ATP
Relative light units
2000
1500
1000
1000
0
Control
LTX-315 5 min
LTX-315 10 min
LTX-315 30 min
LTX-315 60 min
b)
Cytochrome C
ng/ml
10
8
6
4
2
0
c)
rTMSC
Control
5 min
10 min
30 min
60 min
S L S L S L S L S L
HMGB-1

Fig. 2 LTX-315 eradicates treated and non-treated lesions in the three-tumor model

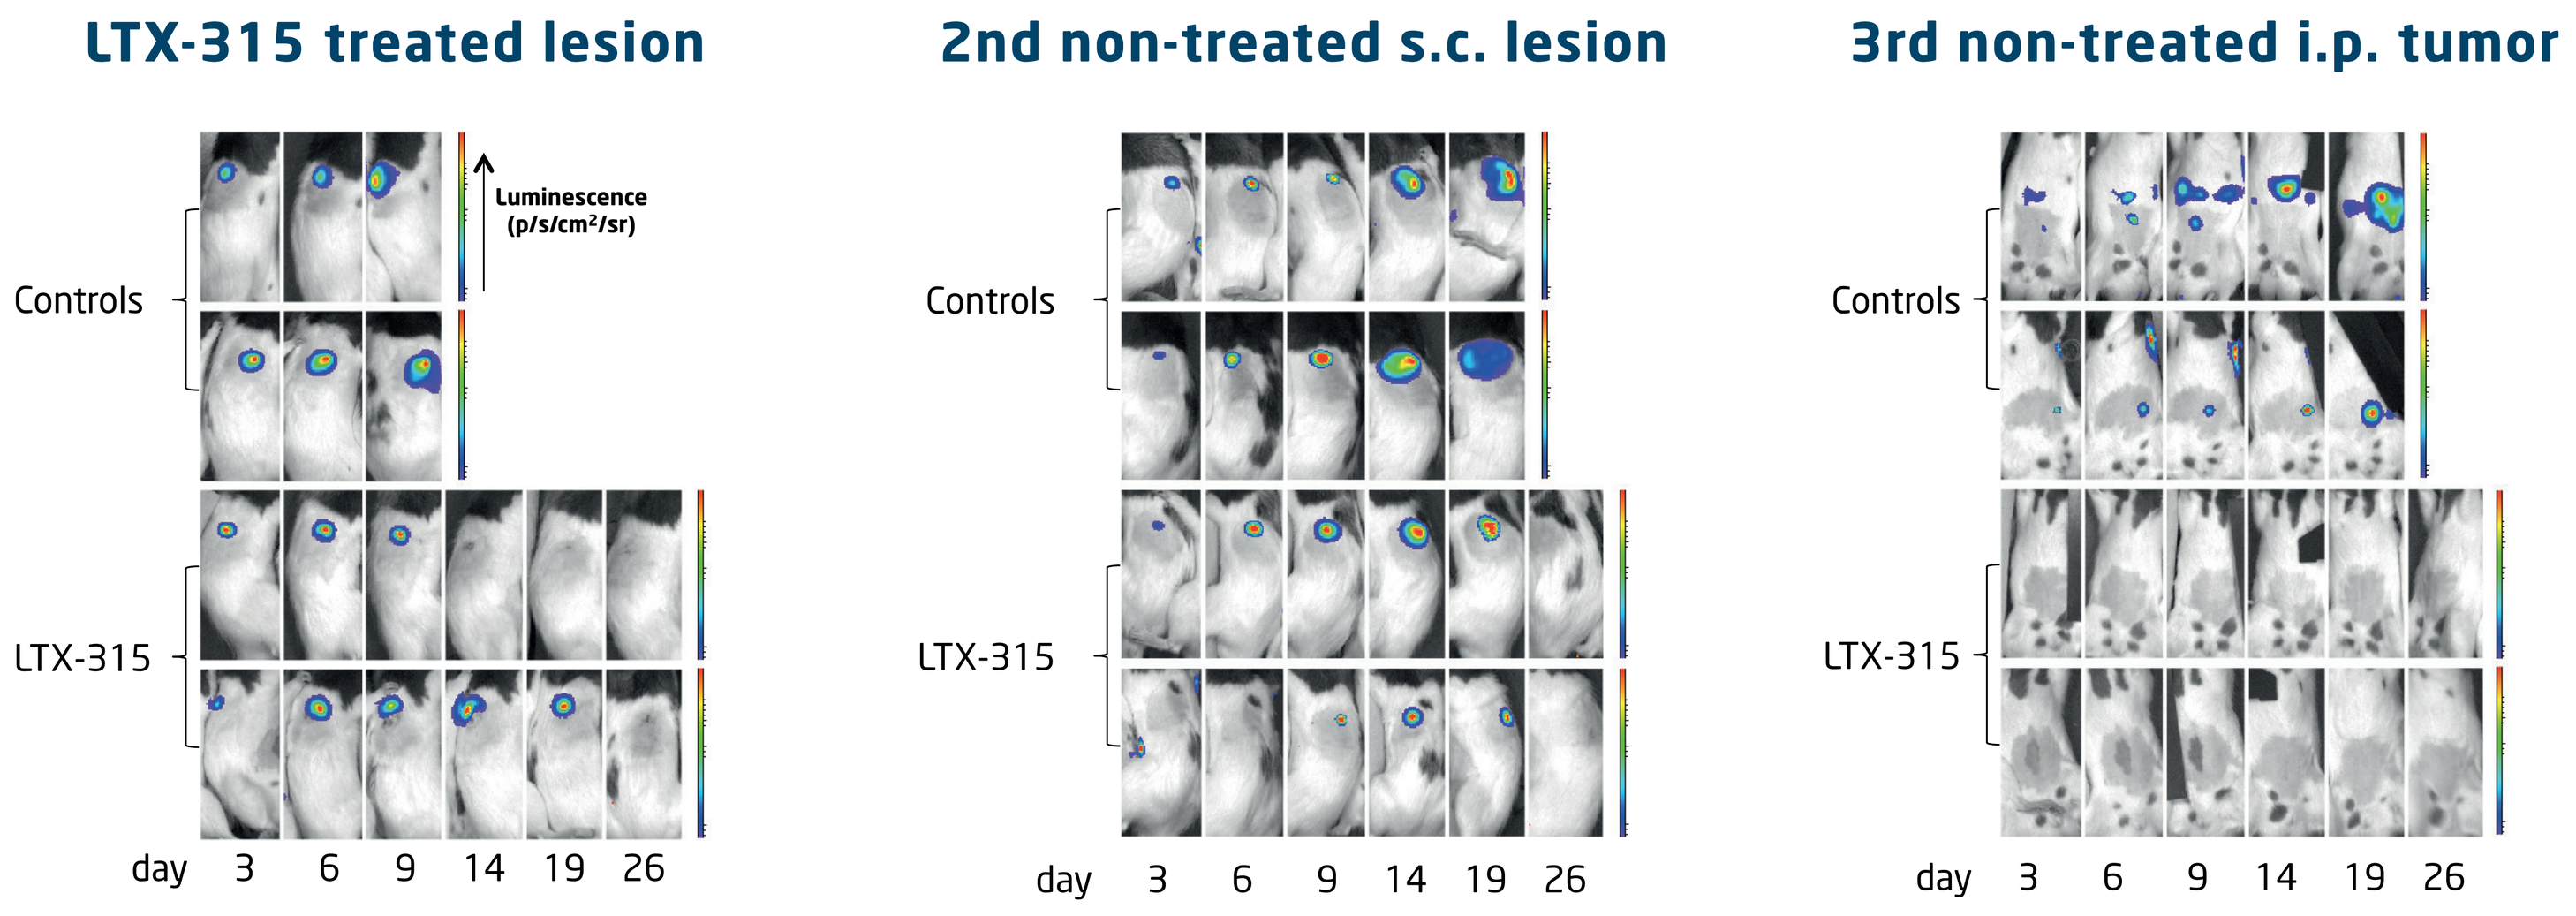
LTX-315 treated lesion
2nd non-treated s.c. lesion
3rd non-treated i.p. tumor
Luminescence (p/s/cm²/sr)
Controls
LTX-315
day 3 6 9 14 19 26

Fig. 3 LTX-315 induces long term protective immune responses

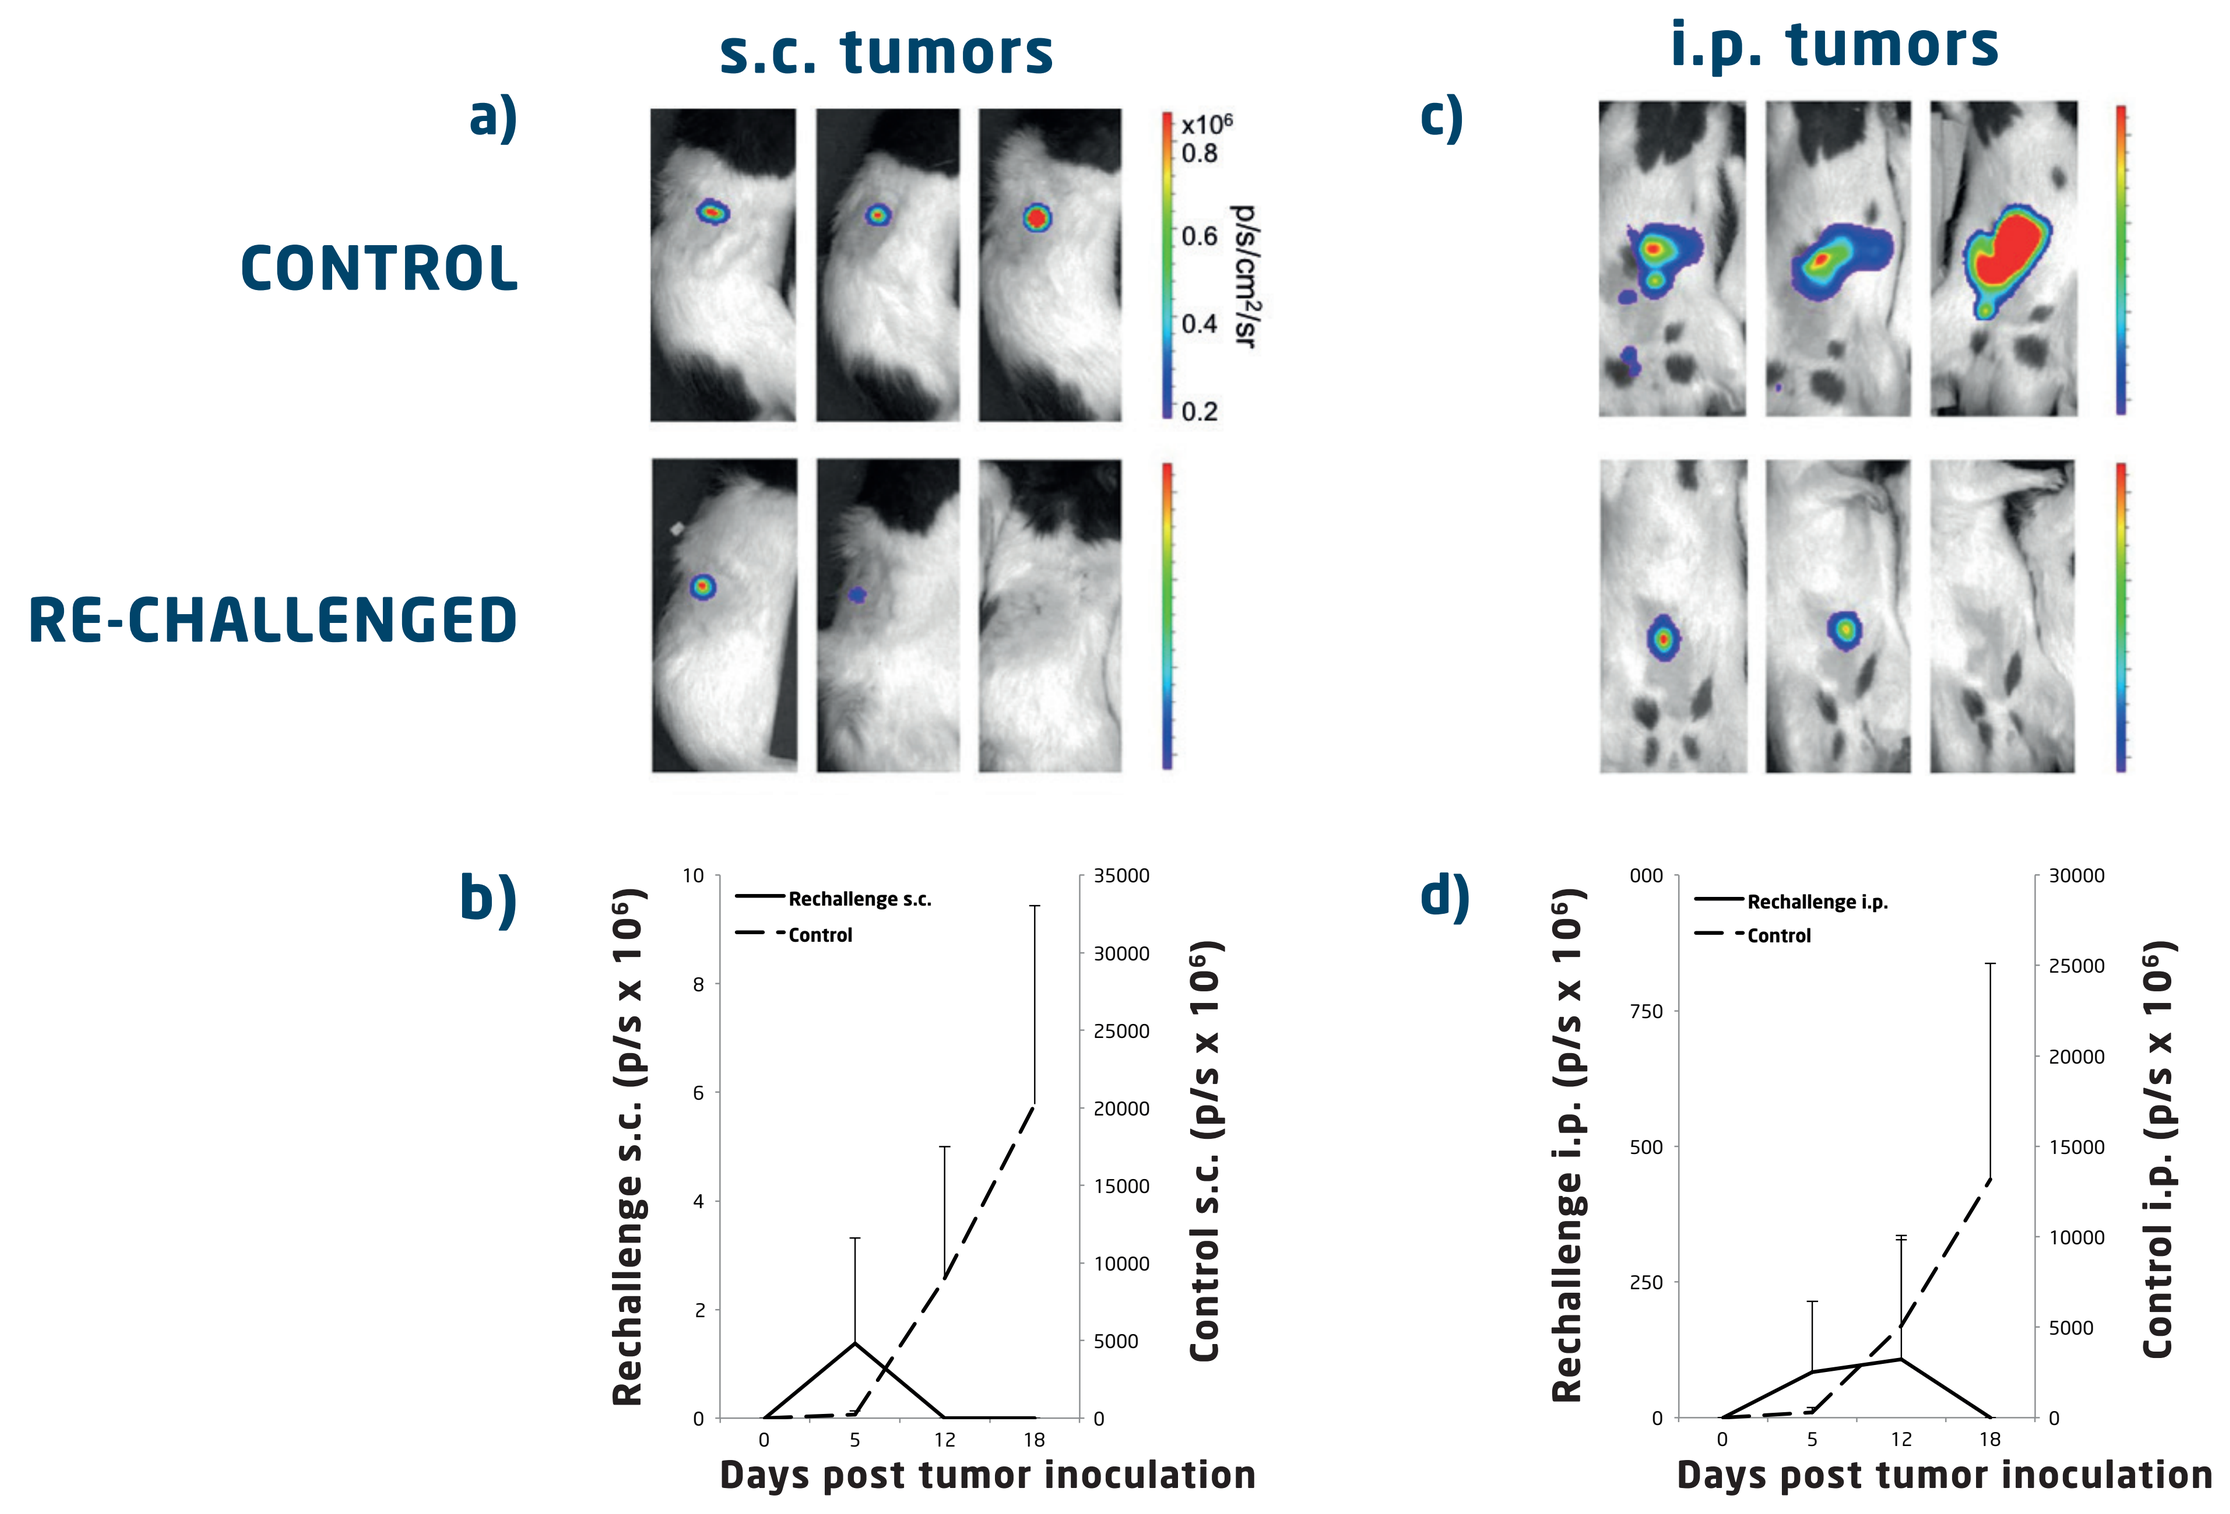
s.c. tumors
i.p. tumors
a)
c)
CONTROL
RE-CHALLENGED
p/s/cm²/sr
b)
d)
Rechallenge s.c. (p/s x 10⁶)
Control s.c. (p/s x 10⁶)
Rechallenge i.p. (p/s x 10⁶)
Control i.p. (p/s x 10⁶)
Rechallenge s.c.
Rechallenge i.p.
Control
Days post tumor inoculation

Fig. 4 Immunohistochemical analysis of LTX-315-treated tumors

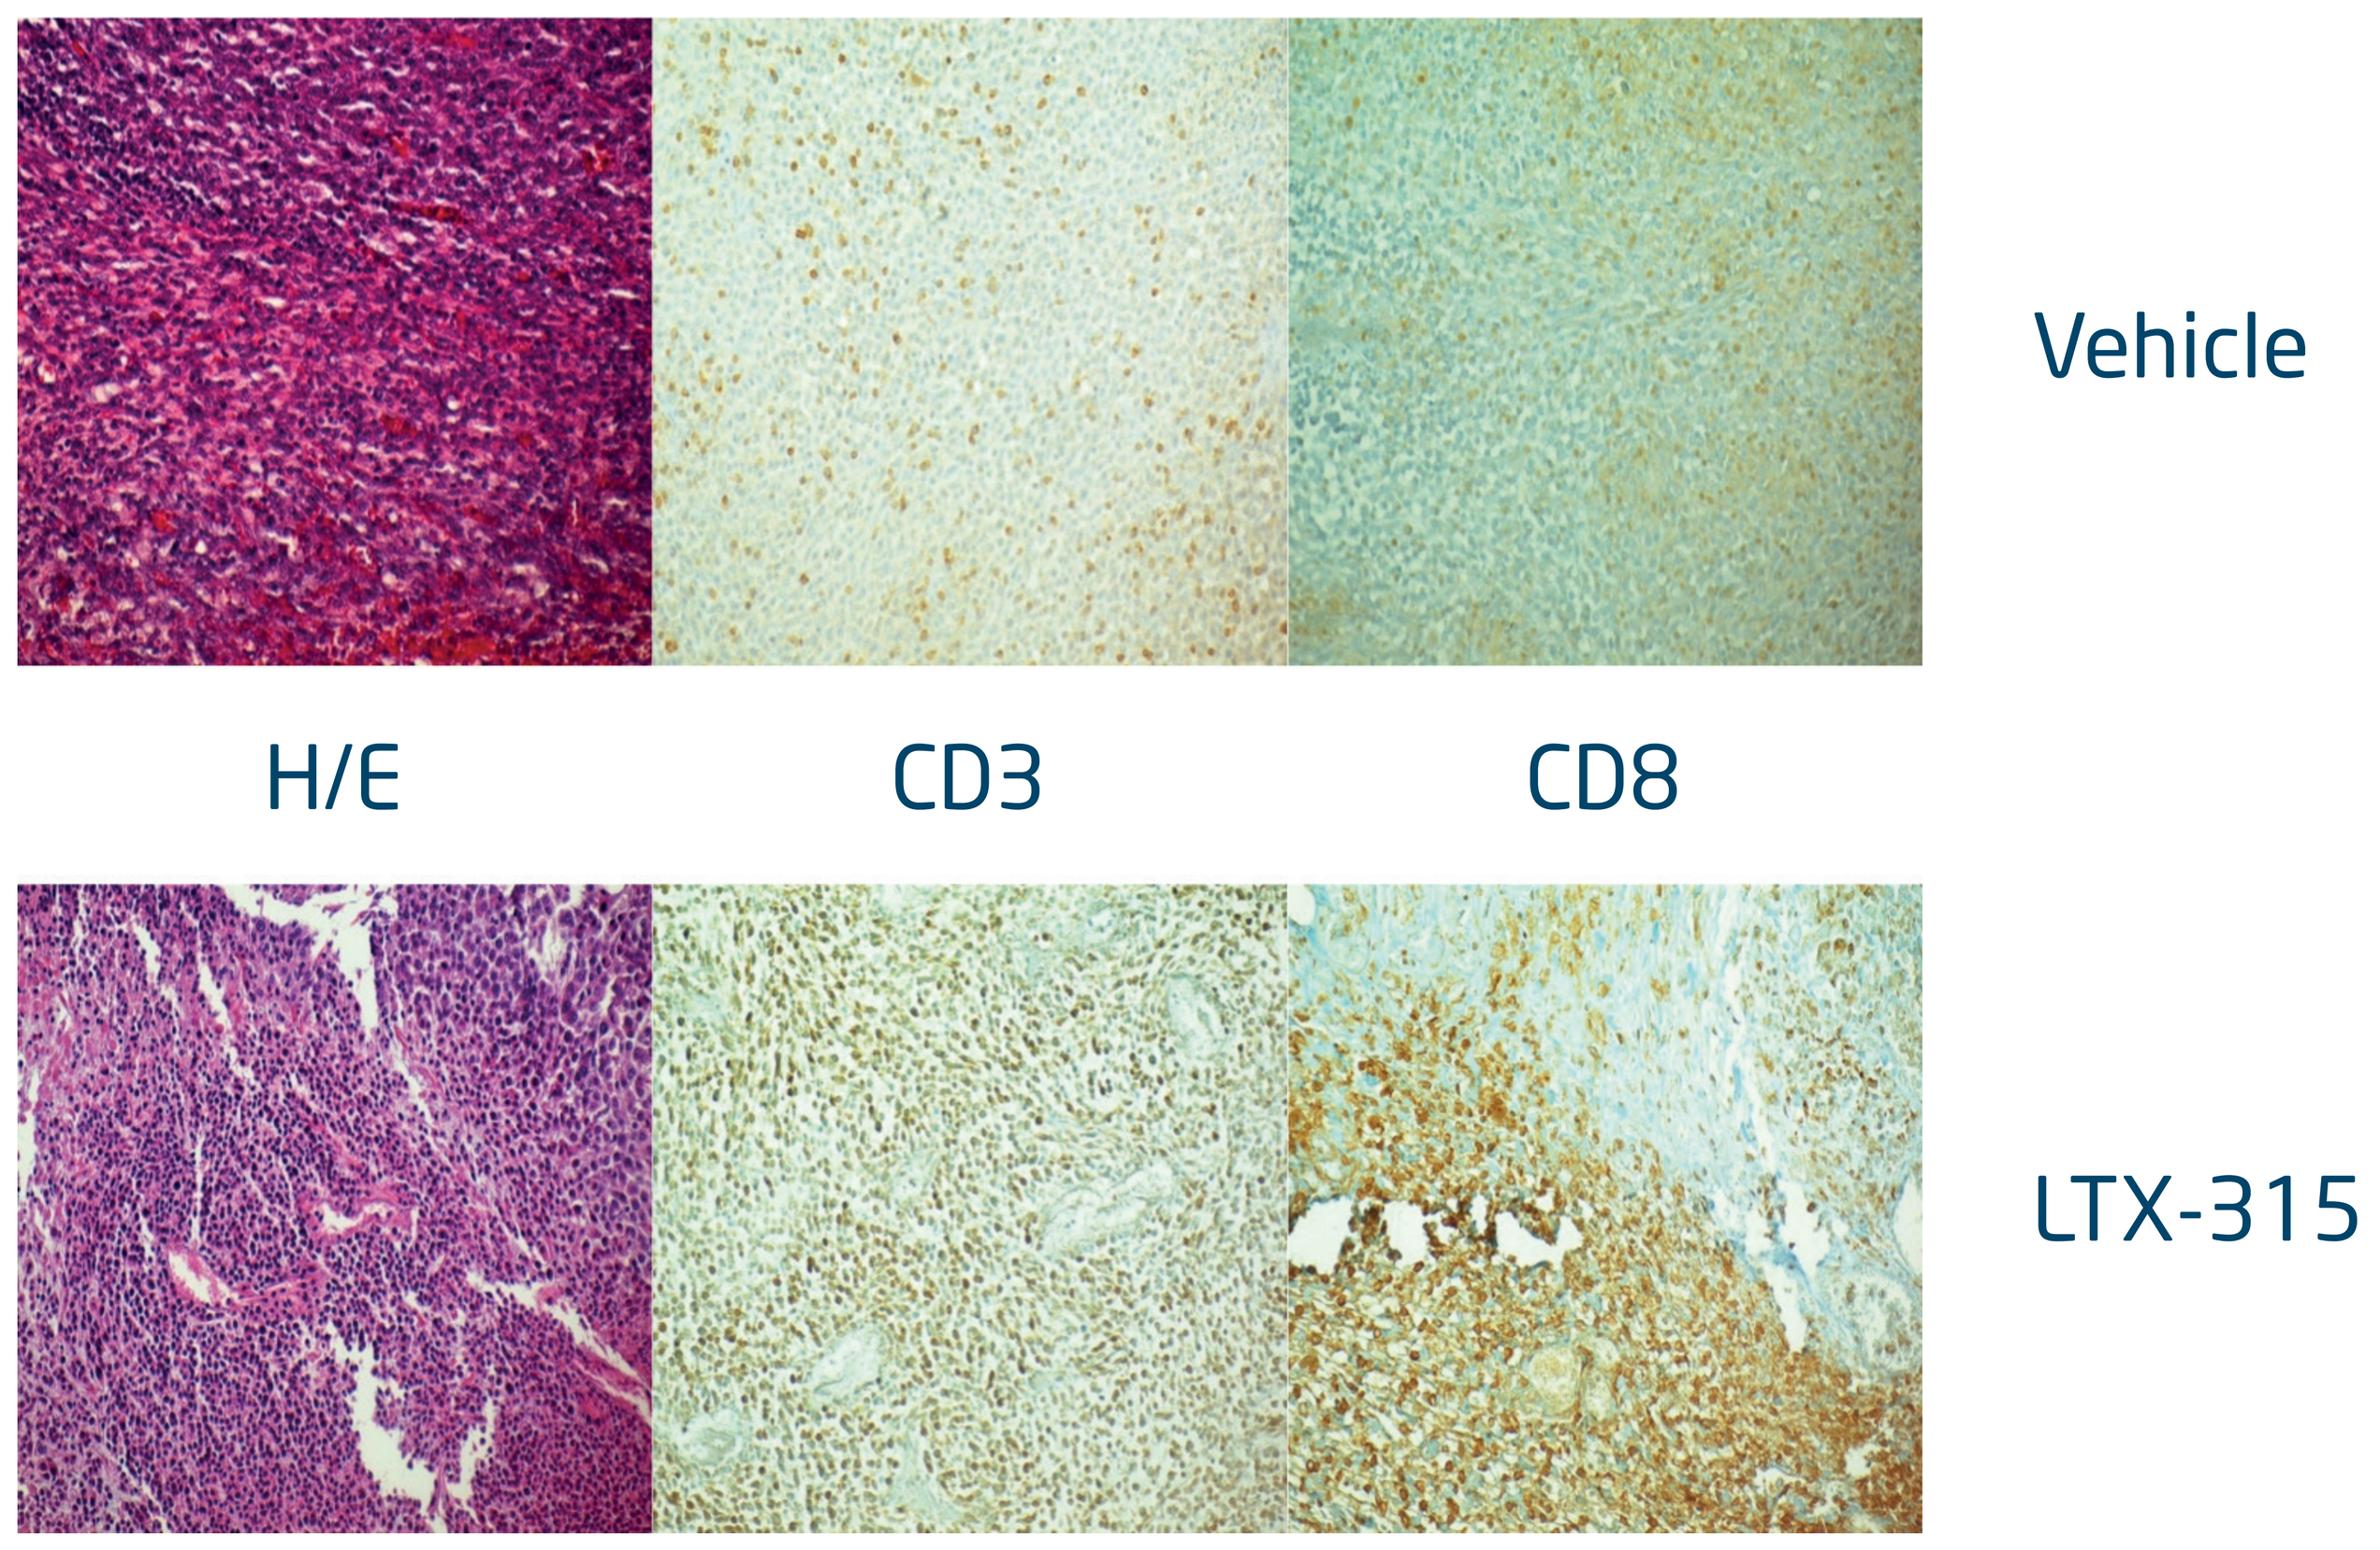
Vehicle
H/E
CD3
CD8
LTX-315

Conclusions

- Here we demonstrate that intralesional treatment of one single lesion with LTX-315 (Oncopore™) is sufficient to cure animals with disseminated tumors.
- Systemic and long lasting protective immune responses were obtained in LTX-315 cured animals.
- LTX-315 induced an increased infiltration of CD3+ and CD8+T cells into the tumor.
- LTX-315 represents a novel intralesional therapeutic strategy with potential to induce clinical responses in metastatic diseases.
- A phase 1/2a study is in progress with LTX-315.

References

1. Berge et al. Cancer Immunol Immunother (2010) 59:1285-1294
2. Camilio et al. Cancer Immunol Immunother (2014) 63:601-13
3. Camilio et al. Oncoimmunology (2014) 25:3

University of Tromsø
Oslo University Hospital
UiO : University of Oslo

© WWW.CHILIGROUP.NO|2014

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: post@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01